Phase I Study on the Safety, Tolerability, and Pharmacodynamic Effects of ASP4345 for the Treatment of Cognitive Impairment in Patients With Schizophrenia

Amit Desai[1], Lauren Benner[1], Ruishan Wu[1], Lev Gertsik[2], Paul Maruff[3], Gregory Light[4], Tolga Uz[1], Gerard Marek[1]

[1]Astellas Pharma Global Development, Inc., Northbrook, IL; [2]California Clinical Trials Medical Group, Inc., Glendale, CA; [3]CogState, Inc., Melbourne, AU; [4]University of California, San Diego, San Diego, CA

Abstract 140

Schizophrenia International Research Society

April 10–14, 2019 | Orlando, FL

BACKGROUND

- Cognitive impairment associated with schizophrenia (CIAS) includes significant deficits in attention/vigilance, speed of processing, working memory, verbal learning, visual learning, reasoning, and problem solving[1,2]
 - This impairment is present in patients with schizophrenia who are on antipsychotics or naive to treatment[1] and currently, no pharmacotherapy has demonstrated efficacy in improving CIAS in clinical trials[3]
- Stimulation of the dopamine type 1 (D_1) receptors in the dorsolateral prefrontal cortex are potential therapeutic targets for CIAS[4]
- Positive allosteric modulators (PAMs) of D_1 receptors enhance endogenous dopamine activity[5]
 - A selective subclass of D_1 receptor PAMs bind less conserved regions of the receptor resulting in increased binding specificity and lower incidence of off-target binding as compared to orthosteric D_1 receptor agonists
- ASP4345 is a novel, small-molecule PAM that selectively binds to, and enhances the activity of, D_1 receptors
- This is a phase I, placebo-controlled, multiple-ascending dose study (NCT02720263) in patients with schizophrenia or schizoaffective disorder who are stabilized on maintenance therapy
- Here we present the primary objective of the safety and tolerability profile of multiple-ascending dose ASP4345 as well as the exploratory objective of the pharmacodynamic effects of ASP4345 on cognition and neurophysiological biomarkers associated with hearing, decision making, and information processing in patients with stabilized schizophrenia or schizoaffective disorder

METHODS

Study Design and Patient Population

- This double-blinded, randomized, multiple-ascending dose, phase I study (NCT02720263) enrolled adult patients (aged 18–60) with schizophrenia or schizoaffective disorder
 - Diagnosis of schizophrenia or schizoaffective disorder followed the guidelines of the Diagnostic and Statistical Manual of Mental Disorders, 5th edition
 - Patients were considered to have stabilized schizophrenia or schizoaffective disorder if they had low to moderate positive symptom scores and moderate negative symptom scores on the positive and negative syndrome scale (PANSS)
- Patients must have been on stable doses (≤25% change in dose) of risperidone, quetiapine, olanzapine, ziprasidone, brexpiprazole, aripiprazole, paliperidone, or lurasidone ≥2 months for oral formulations, or ≥3 months for depot formulations, prior to screening
- Patients were assigned to one of four dose cohorts and randomized 3:1 within each cohort to receive ASP4345 or placebo
- The first cohort (Cohort A) received 3-mg ASP4345 or placebo; thereafter, dose levels were not fixed
 - Between all cohorts, the sponsor and investigator/delegate decided on the next dose level
- The remaining cohorts received 15-mg (Cohort B), 50-mg (Cohort C), or 150-mg (Cohort D) ASP4345 or placebo
- Treatment was orally administered once daily for 14 days and on Days 1, 7, and 14, administration was done under fasting conditions

Assessments and Statistical Analysis

- Tolerability and safety were based on monitoring of adverse event (AEs) with physical and mental health examinations, clinical laboratory evaluations, motor disorder diagnostics, and electrocardiogram monitoring
 - Mental health evaluations included abuse liability with addiction Research Center Inventory (49-item short form), Columbia-suicide severity rating scale, Bond-Lader visual analogue scale, PANSS, and clinical global impression
 - Movement disorder diagnostics included abnormal involuntary movement scale, Simpson Angus scale, and Barnes Akathisia rating scale to assess tardive dyskinesia, drug-induced neurological effects, and drug-induced akathisia, respectively
- Cognition was measured using the Detection Task, Identification Task, and One Back Task from the Cogstate battery test to assess psychomotor and information processing, visual attention, and attention and working memory, respectively
- Changes to neurophysiological biomarkers were measured by electroencephalogram and electrophysiological measurements for auditory steady-state response (ASSR), mismatch negativity (MMN), and P300 to assess hearing, decision making, and information processing, respectively

RESULTS

Patient Disposition, Baseline Demographics, and Disease Characteristics

- Forty-eight patients with schizophrenia or schizoaffective disorder received placebo (N=12) or ASP4345 (N=36; 3-mg; 15-mg; 50-mg; 150-mg [n=9 each dose])
- The demographics and baseline characteristics are shown in **Table 1**

Table 1. Subject Demographics and Baseline Disease Characteristics of Patients With Schizophrenia Treated With Placebo or ASP4345

Characteristics		Placebo (N=12)	ASP4345 (N=36) 3 mg (n=9)	15 mg (n=9)	50 mg (n=9)	150 mg (n=9)
Median age, years (range)		52 (27–60)	49 (33–60)	53 (30–60)	53 (38–58)	38 (37–50)
Sex, n (%)	Male	8 (67)	7 (78)	9 (100)	7 (78)	7 (78)
	Female	4 (33)	2 (22)	0	2 (22)	2 (22)
Ethnicity, n (%)	Hispanic or Latino	1 (8)	0	2 (22)	0	1 (11)
	Not Hispanic or Latino	11 (92)	9 (100)	7 (78)	9 (100)	8 (89)
Race, n (%)	Caucasian	2 (17)	1 (11)	2 (22)	0	1 (11)
	African American	10 (83)	7 (78)	6 (67)	9 (100)	8 (89)
	Asian	0	1 (11)	0	0	0
	Other	0	0	1 (11)	0	0
Median weight, kg (range)		86 (70–109)	81 (71–111)	82 (68–137)	86 (54–109)	92 (65–116)
Median height, cm (range)		173 (168–196)	169 (157–184)	175 (168–190)	172 (155–185)	175 (162–183)
BMI, kg/m^2 (range)		27 (23–39)	29 (24–34)	26 (21–39)	31 (19–39)	30 (21–39)
COMT Genotype, n (%)	Met/Met	1 (8)	1 (11)	2 (22)	2 (22)	1 (11)
	Val/Met	6 (50)	4 (44)	3 (33)	3 (33)	6 (67)
	Val/Val	5 (42)	4 (44)	4 (44)	4 (44)	2 (22)

Abbreviations: BMI, body mass index; COMT, catechol-o-methyltransferase; Met, methionine; Val, valine.

Safety and Tolerability

- Of the 48 treated patients, 38 (79%; ASP4345, n=29; placebo, n=9) experienced ≥1 treatment-emergent AE (TEAE)
- Headache and somnolence were the most commonly reported TEAEs in both ASP4345- and placebo-treated patients (**Table 2**)
- Adverse events considered at least possibly related to ASP4345, as assessed by the investigator, occurring in ≥2 patients were constipation (14%, n=5), somnolence (19%, n=7), headache (14%, n=5), abnormal dreams (6%, n=2), anxiety (6%, n=2), and restlessness (6%, n=2)
- The majority of reported AEs, regardless of attribution, were considered mild in severity by the investigator, with the exception of psychotic disorder (n=1; 3-mg ASP4345) and arthralgia (n=1; placebo), both of which were considered moderate in severity, and a small intestinal obstruction (n=1; 50-mg ASP4345), which was considered severe and unrelated to ASP4345
- No considerable difference in AEs were observed between ASP4345 doses or between ASP4345 and placebo
- There were no AEs leading to ASP4345 withdrawal and no deaths were reported throughout the duration of the study

Table 2. Incidence of Treatment-Emergent AEs Occurring in ≥2 Patients in Any Cohort

	Placebo (N=12)	ASP4345 (N=36) 3 mg (n=9)	15 mg (n=9)	50 mg (n=9)	150 mg (n=9)
Headache	4 (33)	0	3 (33)	3 (33)	3 (33)
Somnolence	1 (8)	2 (22)	1 (11)	2 (22)	2 (22)
Constipation	0	2 (22)	0	3 (33)	0
Back pain	0	1 (11)	2 (22)	0	1 (11)
Dyspepsia	1 (8)	0	1 (11)	2 (22)	0
Upper respiratory tract infection	1 (8)	2 (22)	1 (11)	0	0
Insomnia	3 (25)	1 (11)	1 (11)	1 (11)	0
Anxiety	0	0	0	0	2 (22)
Dizziness	2 (17)	1 (11)	1 (11)	0	0
Abnormal dreams	0	2 (22)	0	0	0

All data presented as n (%).
Adverse events listed are individual preferred terms.

- There was no clear evidence that ASP4345 has an abuse liability
 - No or very small median changes from baseline across time points and treatment groups; these observed changes from baseline were considered to be within the range of normal variation and do not indicate any trends towards dose-related impairment
- No single event of suicidal ideation or suicidal behavior was reported that would suggest that ASP4345 has a potential effect on (causing/triggering) suicidal ideation or behavior

Pharmacodynamic Effects on Cognition

- Improvement in Detection and Identification Task scores were observed across ASP4345 treatment groups on Day 14 relative to placebo (**Table 3**)
 - Detection Task score improvements declined between 60% and 80% in effect size after 72 hours of drug withdrawal (Day 17)
 - Identification Task scores declined between 30% and 125% in effect size in the 3-mg, 50-mg, and 150-mg ASP4345 cohorts after 72 hours of drug withdrawal
- The One Back Task scores did not show any trends in improvement during ASP4345 treatment compared with placebo

Table 3. Effect Size Changes in Cognitive Testing Scores Between Placebo and ASP4345 on Day 14 and Day 17

		ASP4345 3 mg (n=9)[a]	15 mg (n=9)	50 mg (n=9)	150 mg (n=9)[b]
Detection Task	Effect size on Day 14	0.820	0.577	0.993	0.589
	Effect size on Day 17[c]	0.264	0.213	0.343	0.135
Identification Task	Effect size on Day 14	0.253	0.426	0.304	0.137
	Effect size on Day 17[c]	0.129	0.352	0.204	-0.034

All data presented as effect size.
[a]n=8 on Days 14 and 17; [b]n=8 on Day 17; [c]72 hours after last dose.

Pharmacodynamic Effects on Neurophysiologic Biomarkers

- Increasing effect sizes for ASSR were observed in patients treated with increasing doses of ASP4345, except 50-mg, by Day 14 (**Figure 1**)
- Small to moderate effect sizes for MMN measurements were observed in patients treated with all doses of ASP4345, except 50-mg, by Day 7 (**Figure 2**)
- There were no considerable improvements in P300 effects compared with placebo (data not shown)

Figure 1. Effect Size Difference Over Time for Auditory Steady-State Response Measurements for Patients Treated With ASP4345 Relative to Placebo

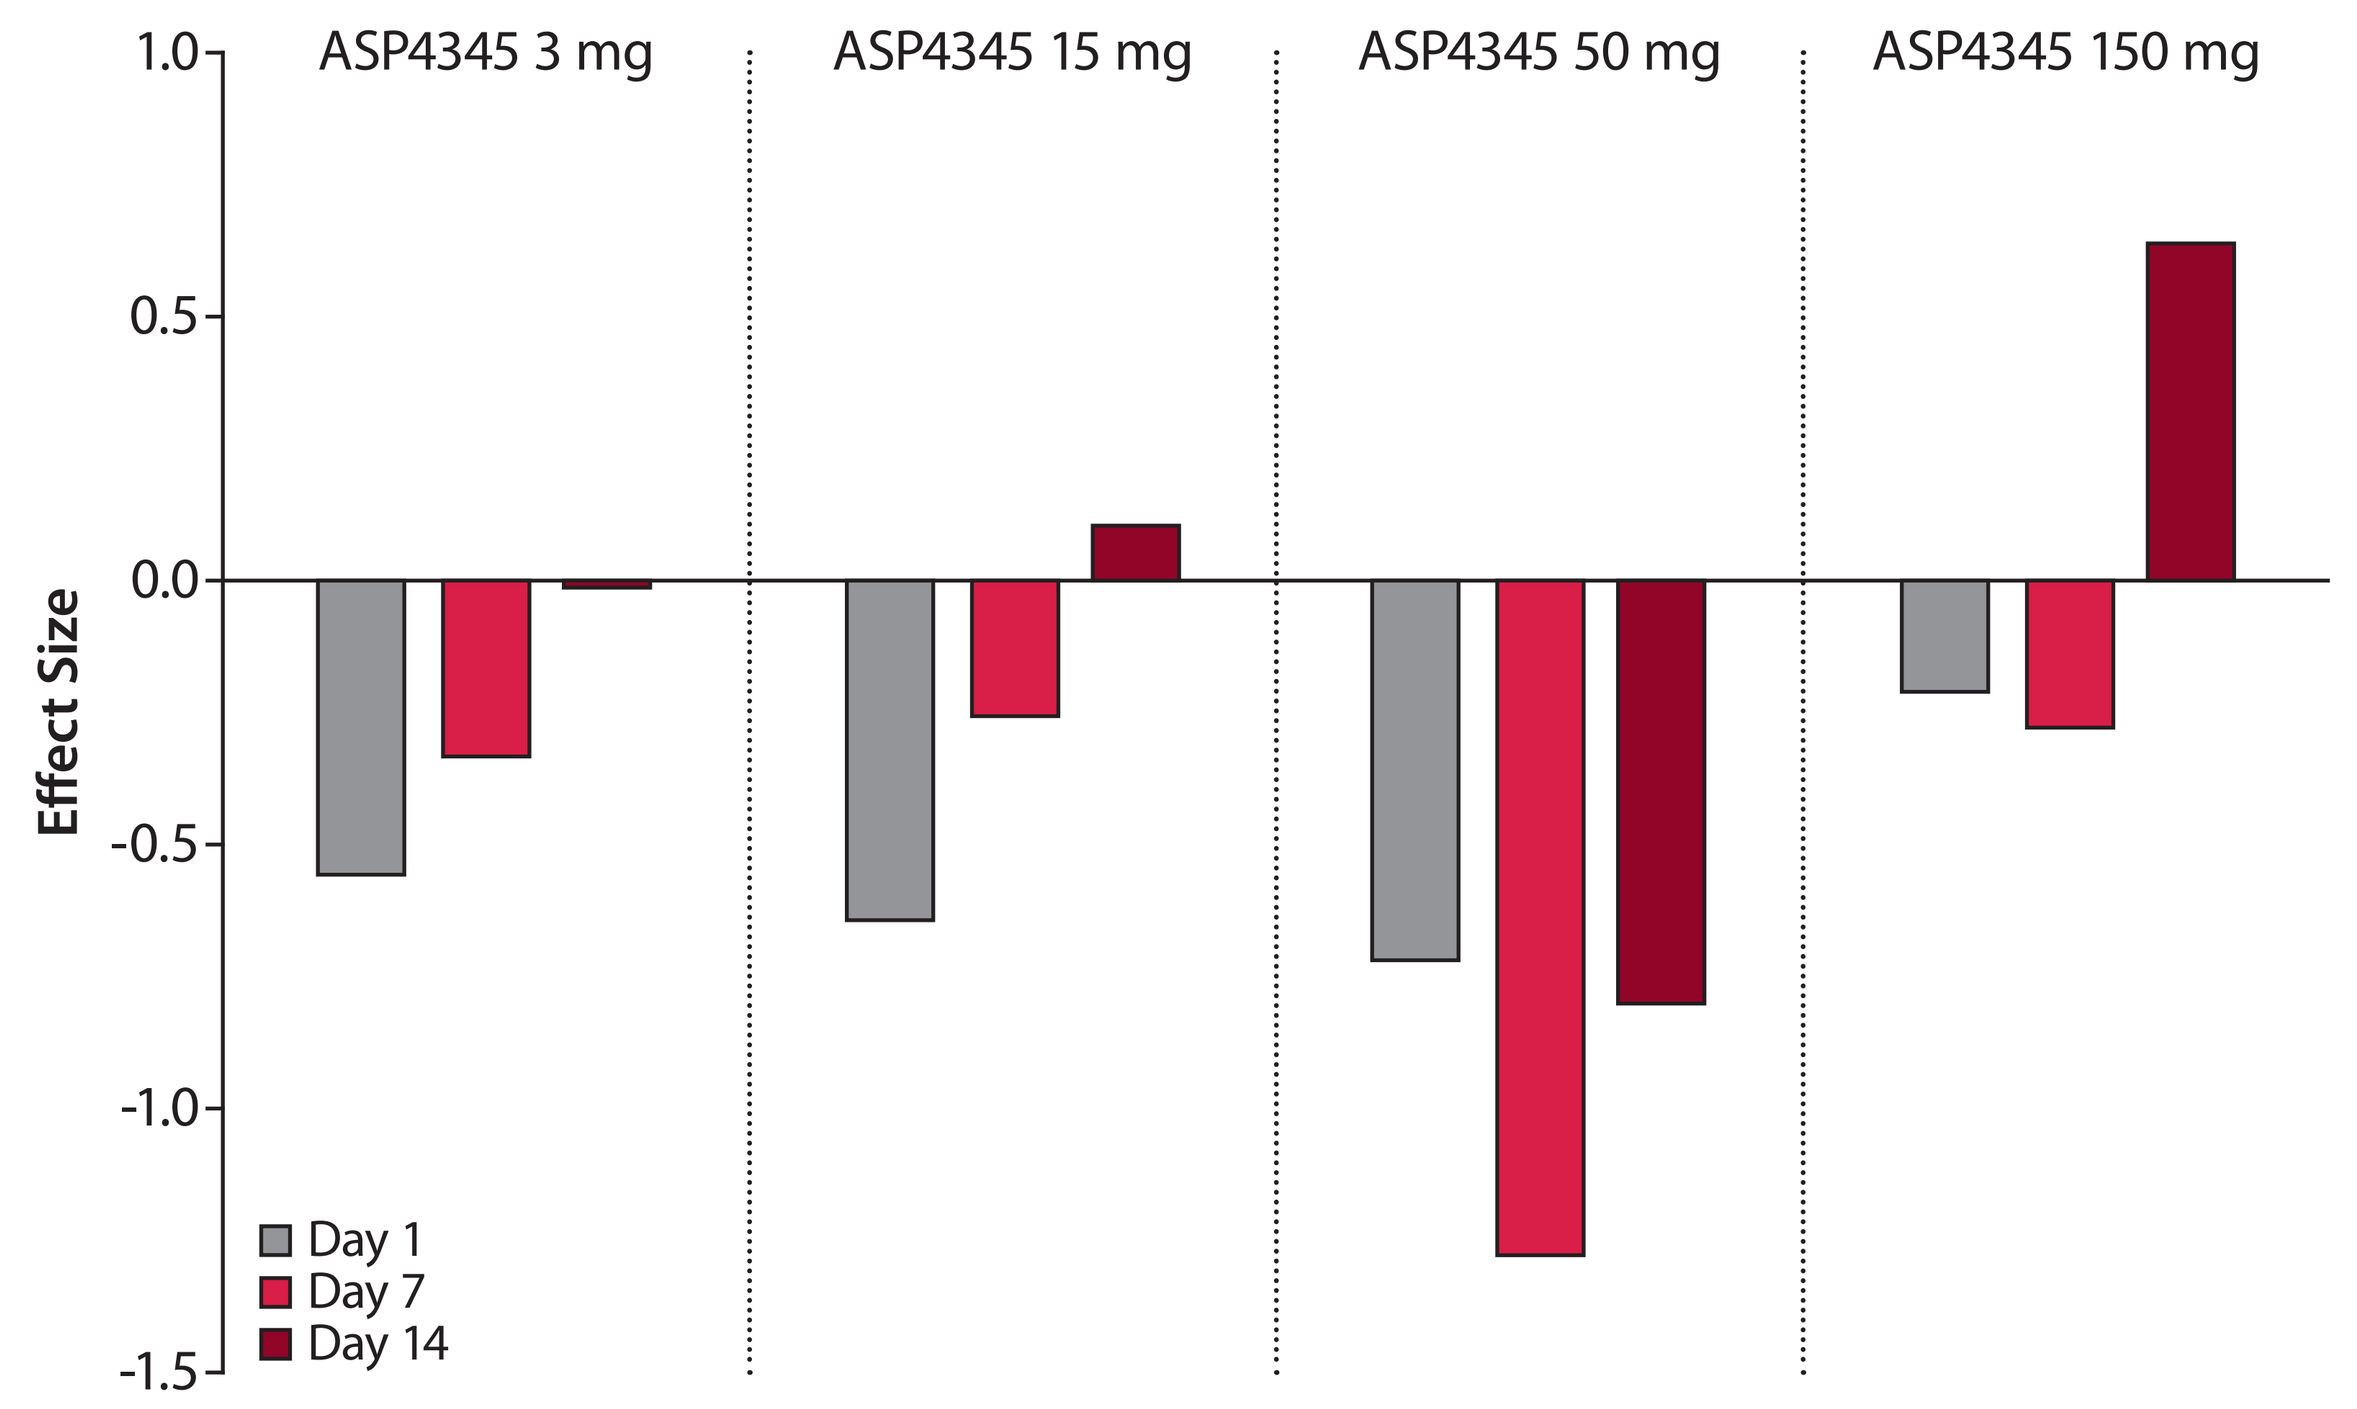
ASP4345 3 mg
ASP4345 15 mg
ASP4345 50 mg
ASP4345 150 mg
Effect Size
1.0
0.5
0.0
-0.5
-1.0
-1.5
Day 1
Day 7
Day 14

Figure 2. Effect Size Difference Over Time for Mismatch Negativity Measurements for Patients Treated With ASP4345 Relative to Placebo

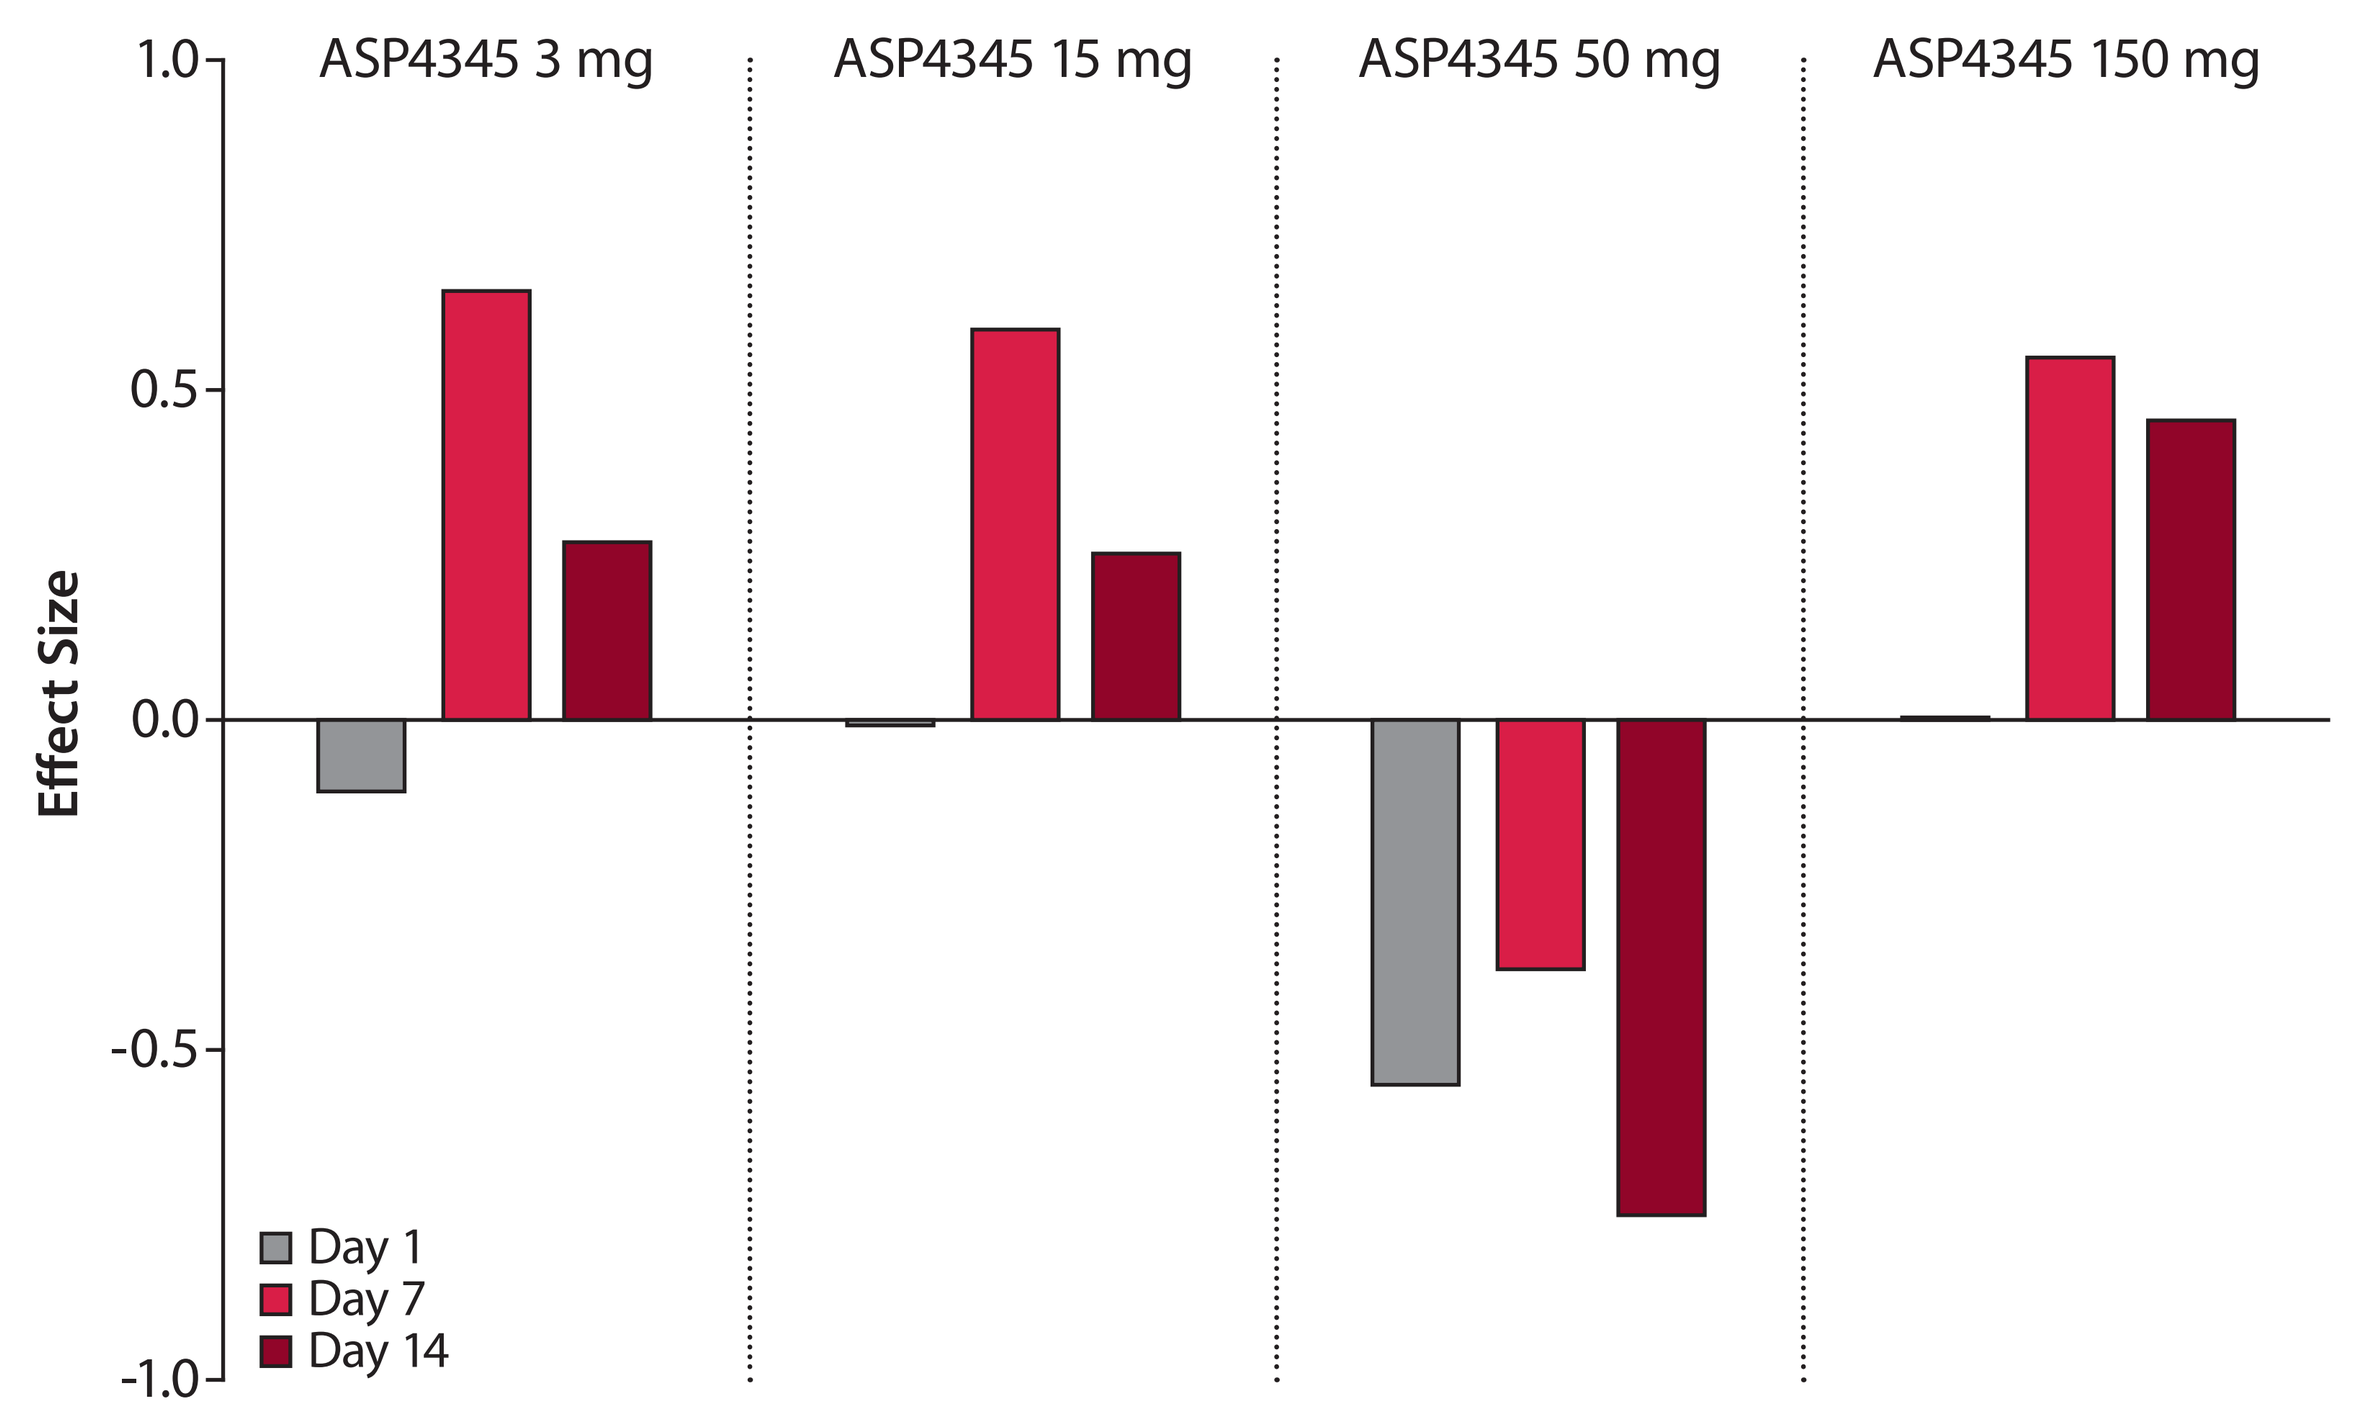
ASP4345 3 mg
ASP4345 15 mg
ASP4345 50 mg
ASP4345 150 mg
Effect Size
1.0
0.5
0.0
-0.5
-1.0
Day 1
Day 7
Day 14

CONCLUSIONS

- All doses of ASP4345 (3-mg, 15-mg, 50-mg, 150-mg) were found to be well tolerated with mostly mild TEAEs and drug-related AEs of constipation, headache, and somnolence
 - There were no signs of abuse liability or effect on mood that could lead to suicidal ideation or behavior
- Improvements in performance on the cognitive tests suggest that treatment with ASP4345 was associated with a benefit in psychomotor function, information processing, and visual attention
- Positive changes in neurophysiological biomarkers ASSR and MMN in patients with schizophrenia and schizoaffective disorder portend improvement in hearing and decision making
- The results of this study highlight the importance of further development of ASP4345 for the treatment of CIAS in patients with schizophrenia and schizoaffective disorder

References

1. Fatouros-Bergman H, Cervenka S, Flyckt L, Edman G, Farde L. Meta-analysis of cognitive performance in drug-naive patients with schizophrenia. *Schizophr Res.* 2014;158:156–162.
2. Schaefer J, Giangrande E, Weinberger DR, Dickinson D. The global cognitive impairment in schizophrenia: consistent over decades and around the world. *Schizophr Res.* 2013;150:42–50.
3. Keefe RS, Buchanan RW, Marder SR, et al. Clinical trials of potential cognitive-enhancing drugs in schizophrenia: what have we learned so far? *Schizophr Bull.* 2013;39:417–435.
4. Tamminga CA. The neurobiology of cognition in schizophrenia. *J Clin Psychiatry.* 2006;67 (suppl 9):9–13; discussion 36–42.
5. Luderman KD, Conroy JL, Free RB, et al. Identification of positive allosteric modulators of the D_1 dopamine receptor that act at diverse binding sites. *Mol Pharmacol.* 2018;94:1197–1209.

Acknowledgments

This study was supported by Astellas Pharma Global Development, Inc. Financial support for this presentation, including writing and editorial assistance by Drs Agnieszka Laskowski and Regina Switzer (OPEN Health Medical Communications, Chicago, IL), was provided by Astellas Pharma Global Development, Inc. (Northbrook, IL, USA).

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Please address any questions or comments regarding this poster to Michael Leski, Senior Manager, Global Scientific Publications, Oncology, Astellas Pharma, Inc. 224-205-5822; michael.leski@astellas.com.