Impaired delayed recall on the International Shopping List Task predicts amyloid positivity and longitudinal decline in CDR-SB scores in MCI

Sharon Rosenzweig-Lipson, PhD[1], Richard Mohs, PhD[1], Paul Maruff, PhD[2], Michela Gallagher, PhD[1,3] and Arnold Bakker, PhD[3]

[1] Agenebio, Inc, Baltimore, MD, USA, [2] Cogstate, Ltd., Melbourne, Victoria, Australia and University of Melbourne, [3] Johns Hopkins University, Baltimore, MD, USA

INTRODUCTION

In older adults with mild cognitive impairment (MCI), abnormally high amyloid (Aβ+) predicts progression to Alzheimer's Disease (AD) dementia. Thus pharmacological intervention in these patients could improve long-term outcome.

The HOPE4MCI trial [NCT03486938] will assess the effects of AGB101 [low dose (220mg) levetiracetam Vs. placebo Q.D.] on clinical disease progression defined by change on the Clinical Dementia Rating Scale Sum Of Boxes score (CDR-SOB) over 18 months in 830 patients with MCI due to AD; confirmed by florbetaben PET.

The International Shopping List Task (ISLT) is a verbal list learning and memory test validated for repeated use in different languages and cultural backgrounds and sensitive to the memory impairment that characterizes MCI. It will be used to identify MCI participants potentially appropriate for the HOPE4MCI trial.

The aim of this study was to provide a basis for determining an optimal criterion for abnormal performance on the ISLT by identifying the ISLT score that best predicts Aβ+, as well as the ISLT score that predicts the greatest decline over 18 months on the CDR-SOB.

METHODS

First, sensitivity to the presence of Aβ+ was determined by comparing estimates of sensitivity and specificity for three levels of baseline impairment on the ISLT (Z</=-1, Z<-/=1.25 and Z</=1.5) in 425 patients who met clinical criteria for MCI and for whom Aβ status had been determined by PET (Table 1).

Second, the sensitivity to decline on CDR-SOB over 18 months was determined for the three levels of baseline impairment on ISLT in 158 Aβ+ MCI patients by computing the effect size for change from baseline to 18 months for each (Table 1).

Participants were enrolled in the AIBL study, had data, including the CDR-SOB for at least 2 assessments 18 months apart and had undergone PET Aβ scanning. MCI diagnoses was made by a consensus panel, blinded to Aβ status using Petersen-Winblad criteria. ISLT data was not utilized by the consensus panel.

Table 1: Demographic and clinical characteristics of AIBL MCI samples used to assess Aim 1 & Aim 2

	Aim 1: Sensitivity to Aβ+		Aim 2: Sensitivity to decline in CDR-SOB
	Aβ- MCI	Aβ+ MCI	Aβ+ MCI
N	170	255	158
% Female	42%	64%	62%
% APOE e4	17%*	55%	52%
Age	76.2 (8.6)	79.5 (6.1)	78.2 (7.8)
Premorbid IQ	109.2 (6.4)	109.4 (5.8)	109.12 (5.6)
MMSE	28.3 (0.9)	25.4 (1.6)	25.8 (2.1)
CDR-SB	0.25 (1.5)	0.5 (3)	0.5 (2)
HADS-D	3.4 (2.3)	3.9 (2.9)	3.7 (2.5)
HADS-A	2.7 (2.2)	4.1 (2.3)	4.2 (2.9)

RESULTS. Aim 1: Sensitivity to Aβ+

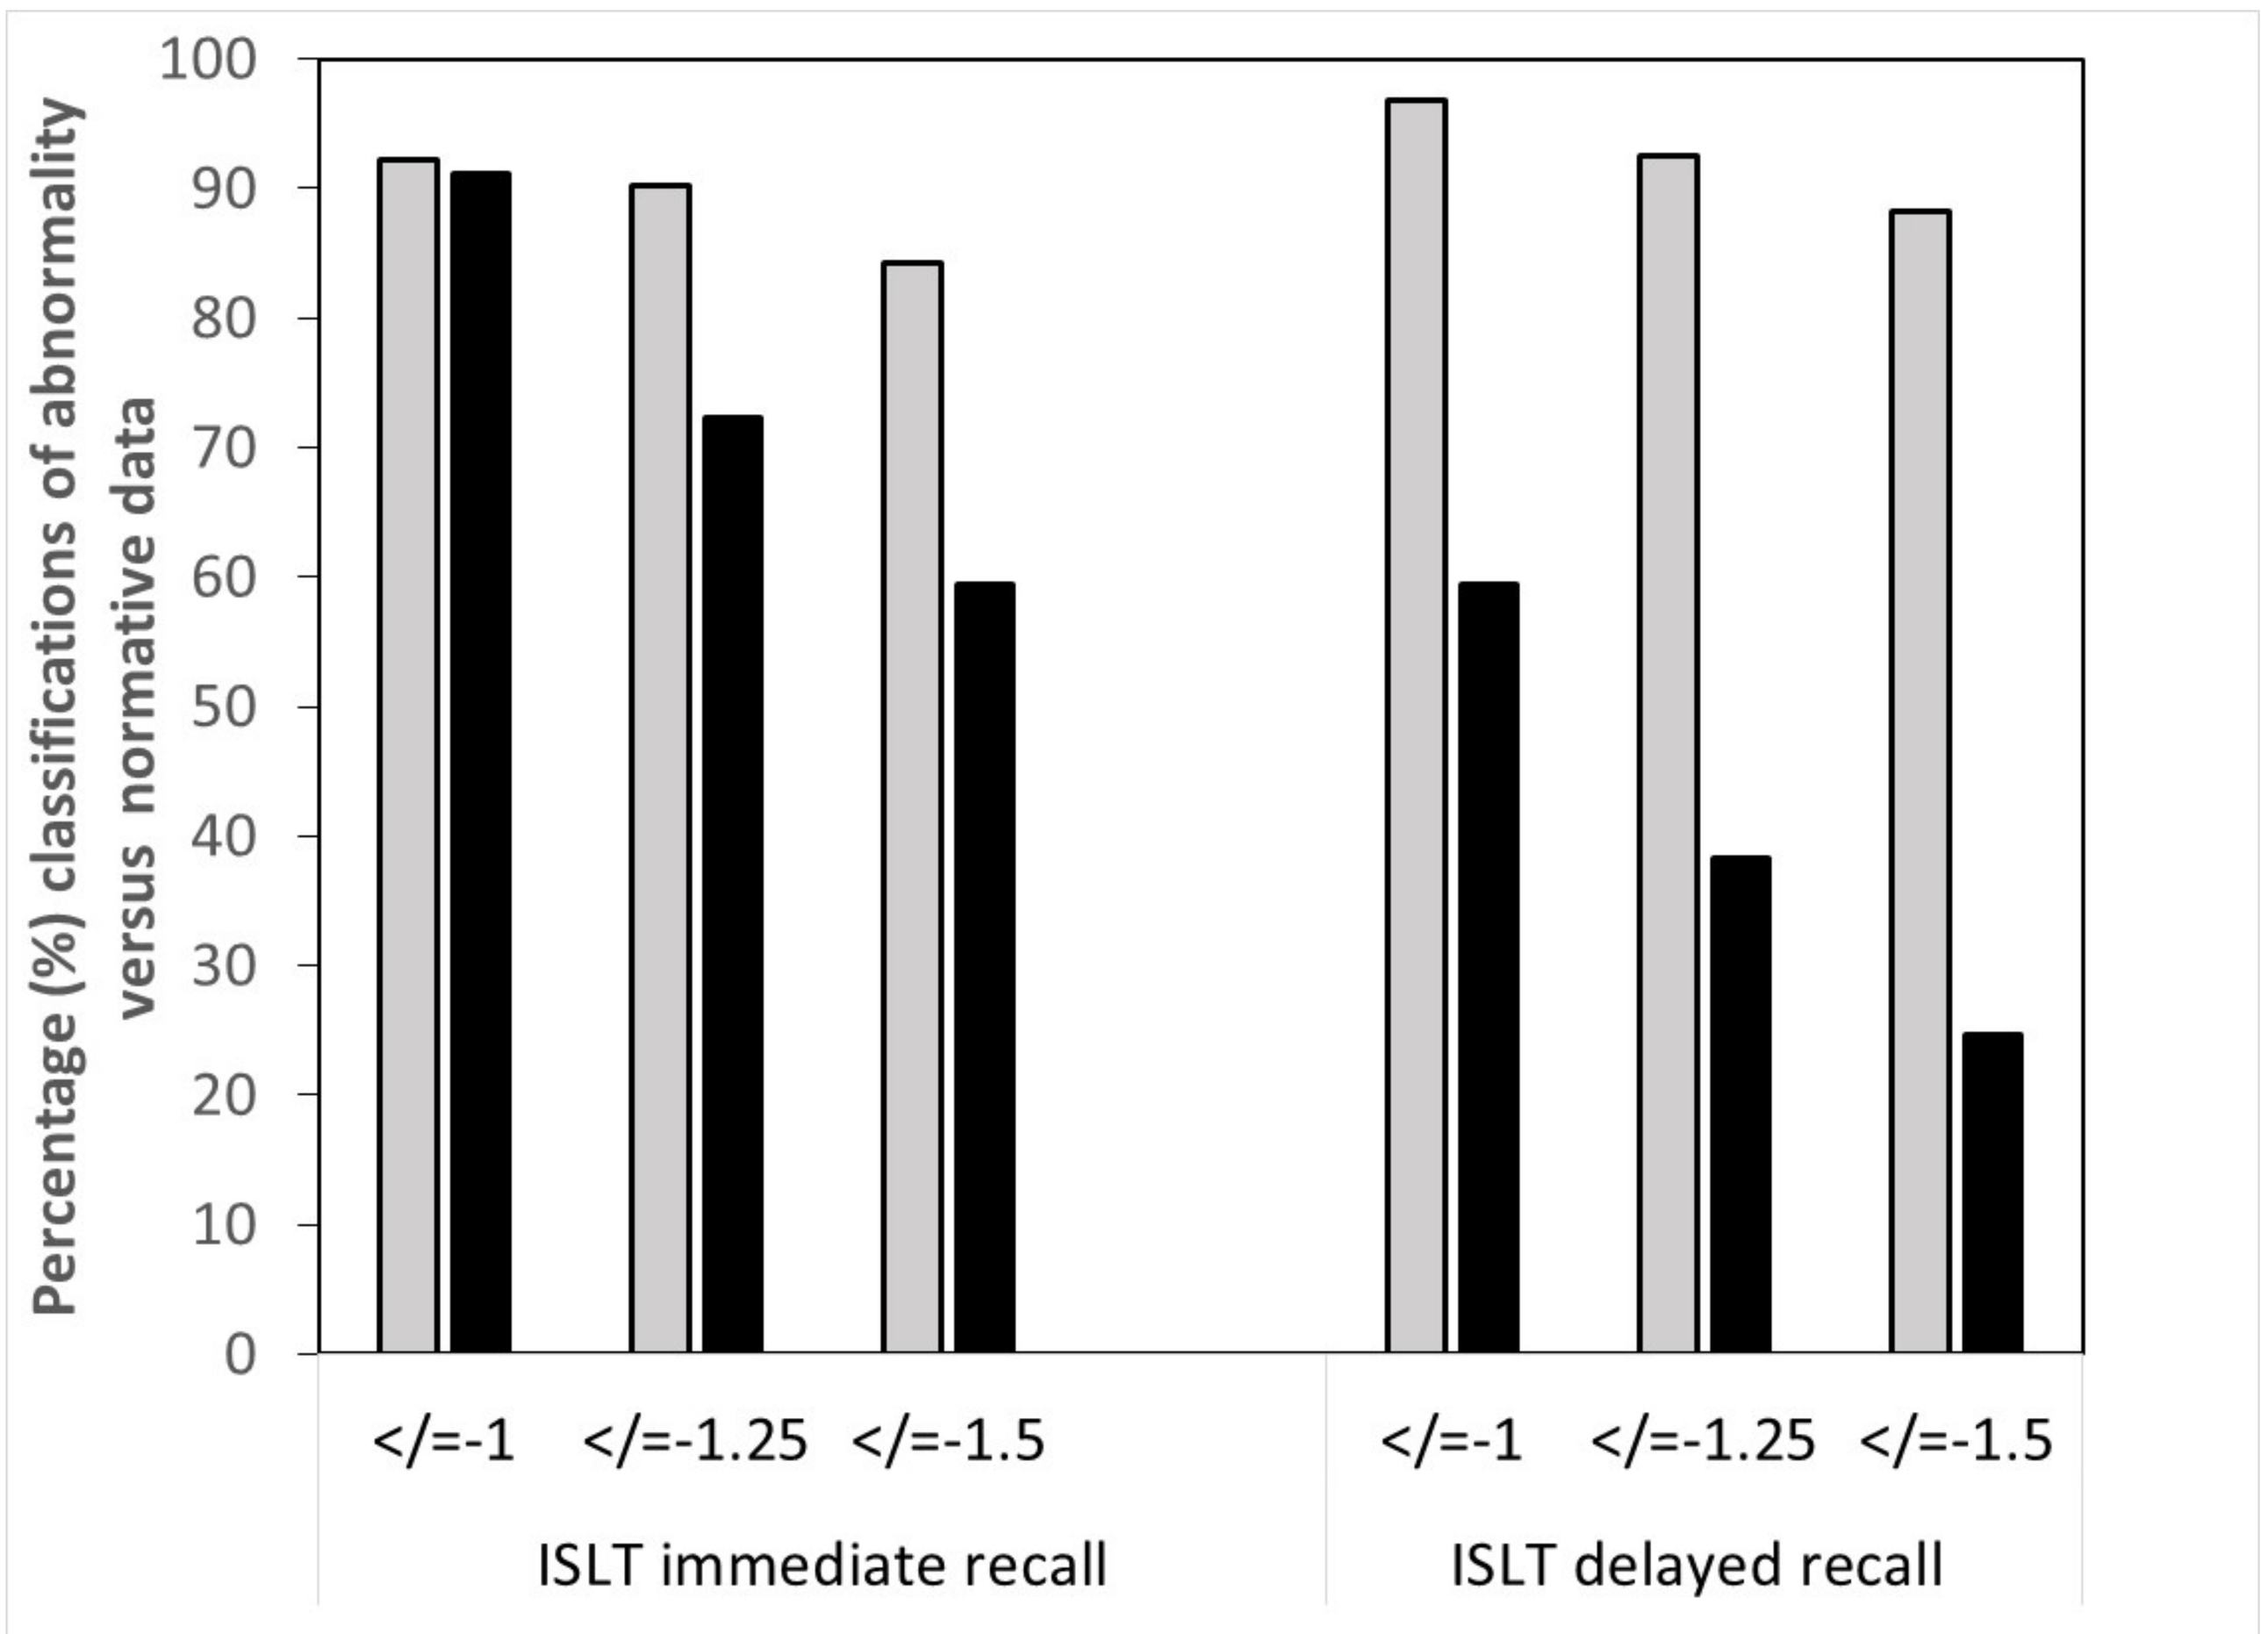
(% Aβ+ abnormal score)
(% Aβ- abnormal score)
Percentage (%) classifications of abnormality versus normative data
100
90
80
70
60
50
40
30
20
10
0
</=-1
</=-1.25
</=-1.5
ISLT immediate recall
ISLT delayed recall

Figure 1: Sensitivity (grey bars) and specificity (black bars) for classification of Aβ+ in older adults with MCI using different cut-scores for abnormality on the ISLT immediate and delayed recall

Figure 1 shows that while each ISLT measures showed high sensitivity to memory impairment in older adults with Aβ+ MCI, the rate of false positive classification was least when the most conservative criteria on the delayed recall trial of the ISLT was used to define abnormality

RESULTS. Aim 2: Sensitivity to CDR decline

Table 2: Number of Aβ+ MCI classified as abnormal for each ISLT cut-score and the associated decline in CDR-SOB score over 18 months

ISLT outcome	Criterion Vs norms	N abnormal	Mean CDR-SOB base-line	Mean CDR-SOB 18 mths	Pooled SD for change in CDR-SOB	Effect size for difference from baseline
Immed-iate recall	<-1	101	1.04	1.9	2.8	**0.31**
	<-1.25	74	1.04	2	2.6	**0.37**
	<-1.5	54	1.06	2.3	2.1	**0.59**
Delayed recall	<-1	131	1.04	1.9	2.3	**0.37**
	<1.25	101	1.23	2.6	1.9	**0.72**
	<-1.5	81	1.48	3.2	1.67	**1.03**

The data in Table 2 show that a greater decline in CDR-SOB at 18 months occurred for Aβ+ MCI patients classified with abnormal memory performance using the ISLT delayed recall score. Furthermore, the magnitude of decline increased to the greatest magnitude when the most conservative criterion was used to classify abnormal memory at baseline.

CONCLUSION

In adults with MCI, definition of memory impairment as performance of </=-1.5 standard deviation units below age matched controls on the delayed recall of the ISLT provided the greatest balance of sensitivity and specificity of selection of Aβ+ in adults with MCI on a single assessment.

The criterion of </=-1.5 standard deviation units below age matched controls on the delayed recall trial of the ISLT was also associated with the greatest decline in CDR-SOB score over the 18 months following baseline.

This relatively conservative criteria for abnormal memory performance in MCI should provide a useful basis for recruitment of individuals with Aβ+ MCI into the HOPE4MCI clinical trial of AGB101.

Acknowledgements AIBL is a collaborative study and a complete list of contributors can be found at www.aibl.csiro.au. The HOPE4MCI trial greatly acknowledge partial support from R01AG048349 to M.G and R56 AG055416 to R.M.